Effects of GM-CSF on the function of polymorphonuclear cells from healthy dogs and dogs with neoplasia

Celia Friedman Cowan, Yan Zhang, Sandra Axiak-Bechtel, Juliana Amorim, Kaoru Tsuruta, Amy DeClue
Comparative Internal Medicine Laboratory, College of Veterinary Medicine, University of Missouri

Introduction

Cancer is the leading cause of death in adult dogs and while chemotherapy is a common treatment, most dogs relapse and die[1,2,3]. Cancer causes immunosuppression and administration of chemotherapy further suppresses the number and function of polymorphonuclear cells (PMNs). PMNs are key in the innate immune system, especially in the recognition of lipopolysaccharide (LPS) of gram-negative bacteria via toll-like receptor 4 (TLR4) and exhibit phagocytic, cytotoxic, and anti-tumor functions. Also, a class of human leukocyte antigen, HLADR, is involved in the adaptive immune response and expressed on macrophages, B cells, and dendritic cells, but can be induced on PMNs[4]. Therefore, dogs with cancer, especially those undergoing chemotherapy, have a greater risk of infection (and reduced anti-tumor cytotoxicity), limiting the amount of chemotherapy treatment that can be used. By increasing function of PMNs, it is possible that more chemotherapy could be tolerated, leading to higher rates of remission and longer survival times. Granulocyte-macrophage colony-stimulating factor (GM-CSF) is a cytokine involved in proliferation and differentiation of blood cells. It also enhances cytotoxic and phagocytic capabilities of PMNs and increases TLR4 and HLADR expression in people. The use of GM-CSF in dogs with cancer could help reduce infection rates and increase anti-tumor cytotoxicity.

Hypothesis

GM-CSF will increase cytokine production of and TLR4 and HLADR expression on healthy canine PMNs and reverse cancer-induced suppression of cytokine production and expression of TLR4 and HLADR on PMNs.

Methods

Sample collection and GM-CSF stimulation

Healthy dogs were selected based on a volunteer basis and met a range of criteria that defined them as “healthy”. Dogs with neoplasia were client-owned dogs selected for having soft tissue sarcoma and used with consent from the presiding clinician and client. Whole blood was collected and stimulated for 2 hours with control, a low concentration of GM-CSF (0.1 μM), or a high concentration of GM-CSF (1 μM).

Cell viability

After stimulation, cells were evaluated for viability using trypan blue exclusion and a hemocytometer. 100 cells were counted twice, noting the number of dead (blue) vs. living cells (clear).

HLADR and TLR4 expression on PMNs

After stimulation, peripheral mononuclear cells (PBMCs) were isolated. The following samples were analyzed using flow cytometry and Summit software:

- Control (unstained PBMCs)
- Non-stimulated and unchallenged (PBMCs stained with TLR4-PE or HLADR-FITC)
- Non-stimulated and challenged (PBMCs challenged with LPS and stained with TLR4-PE and HLADR-FITC)
- Stimulated and challenged (PBMCs stimulated with 0.1 μM and 1 μM GM-CSF, challenged with LPS, and stained with TLR4-PE and HLADR-FITC)

Leukocyte cytokine production capacity

After stimulation, the blood was challenged with PBS (control) or LPS and allowed to incubate for 24 hours. Supernatant TNF, IL-6, and IL-10 were measured using canine specific bead-based multiplex cytokine assays.

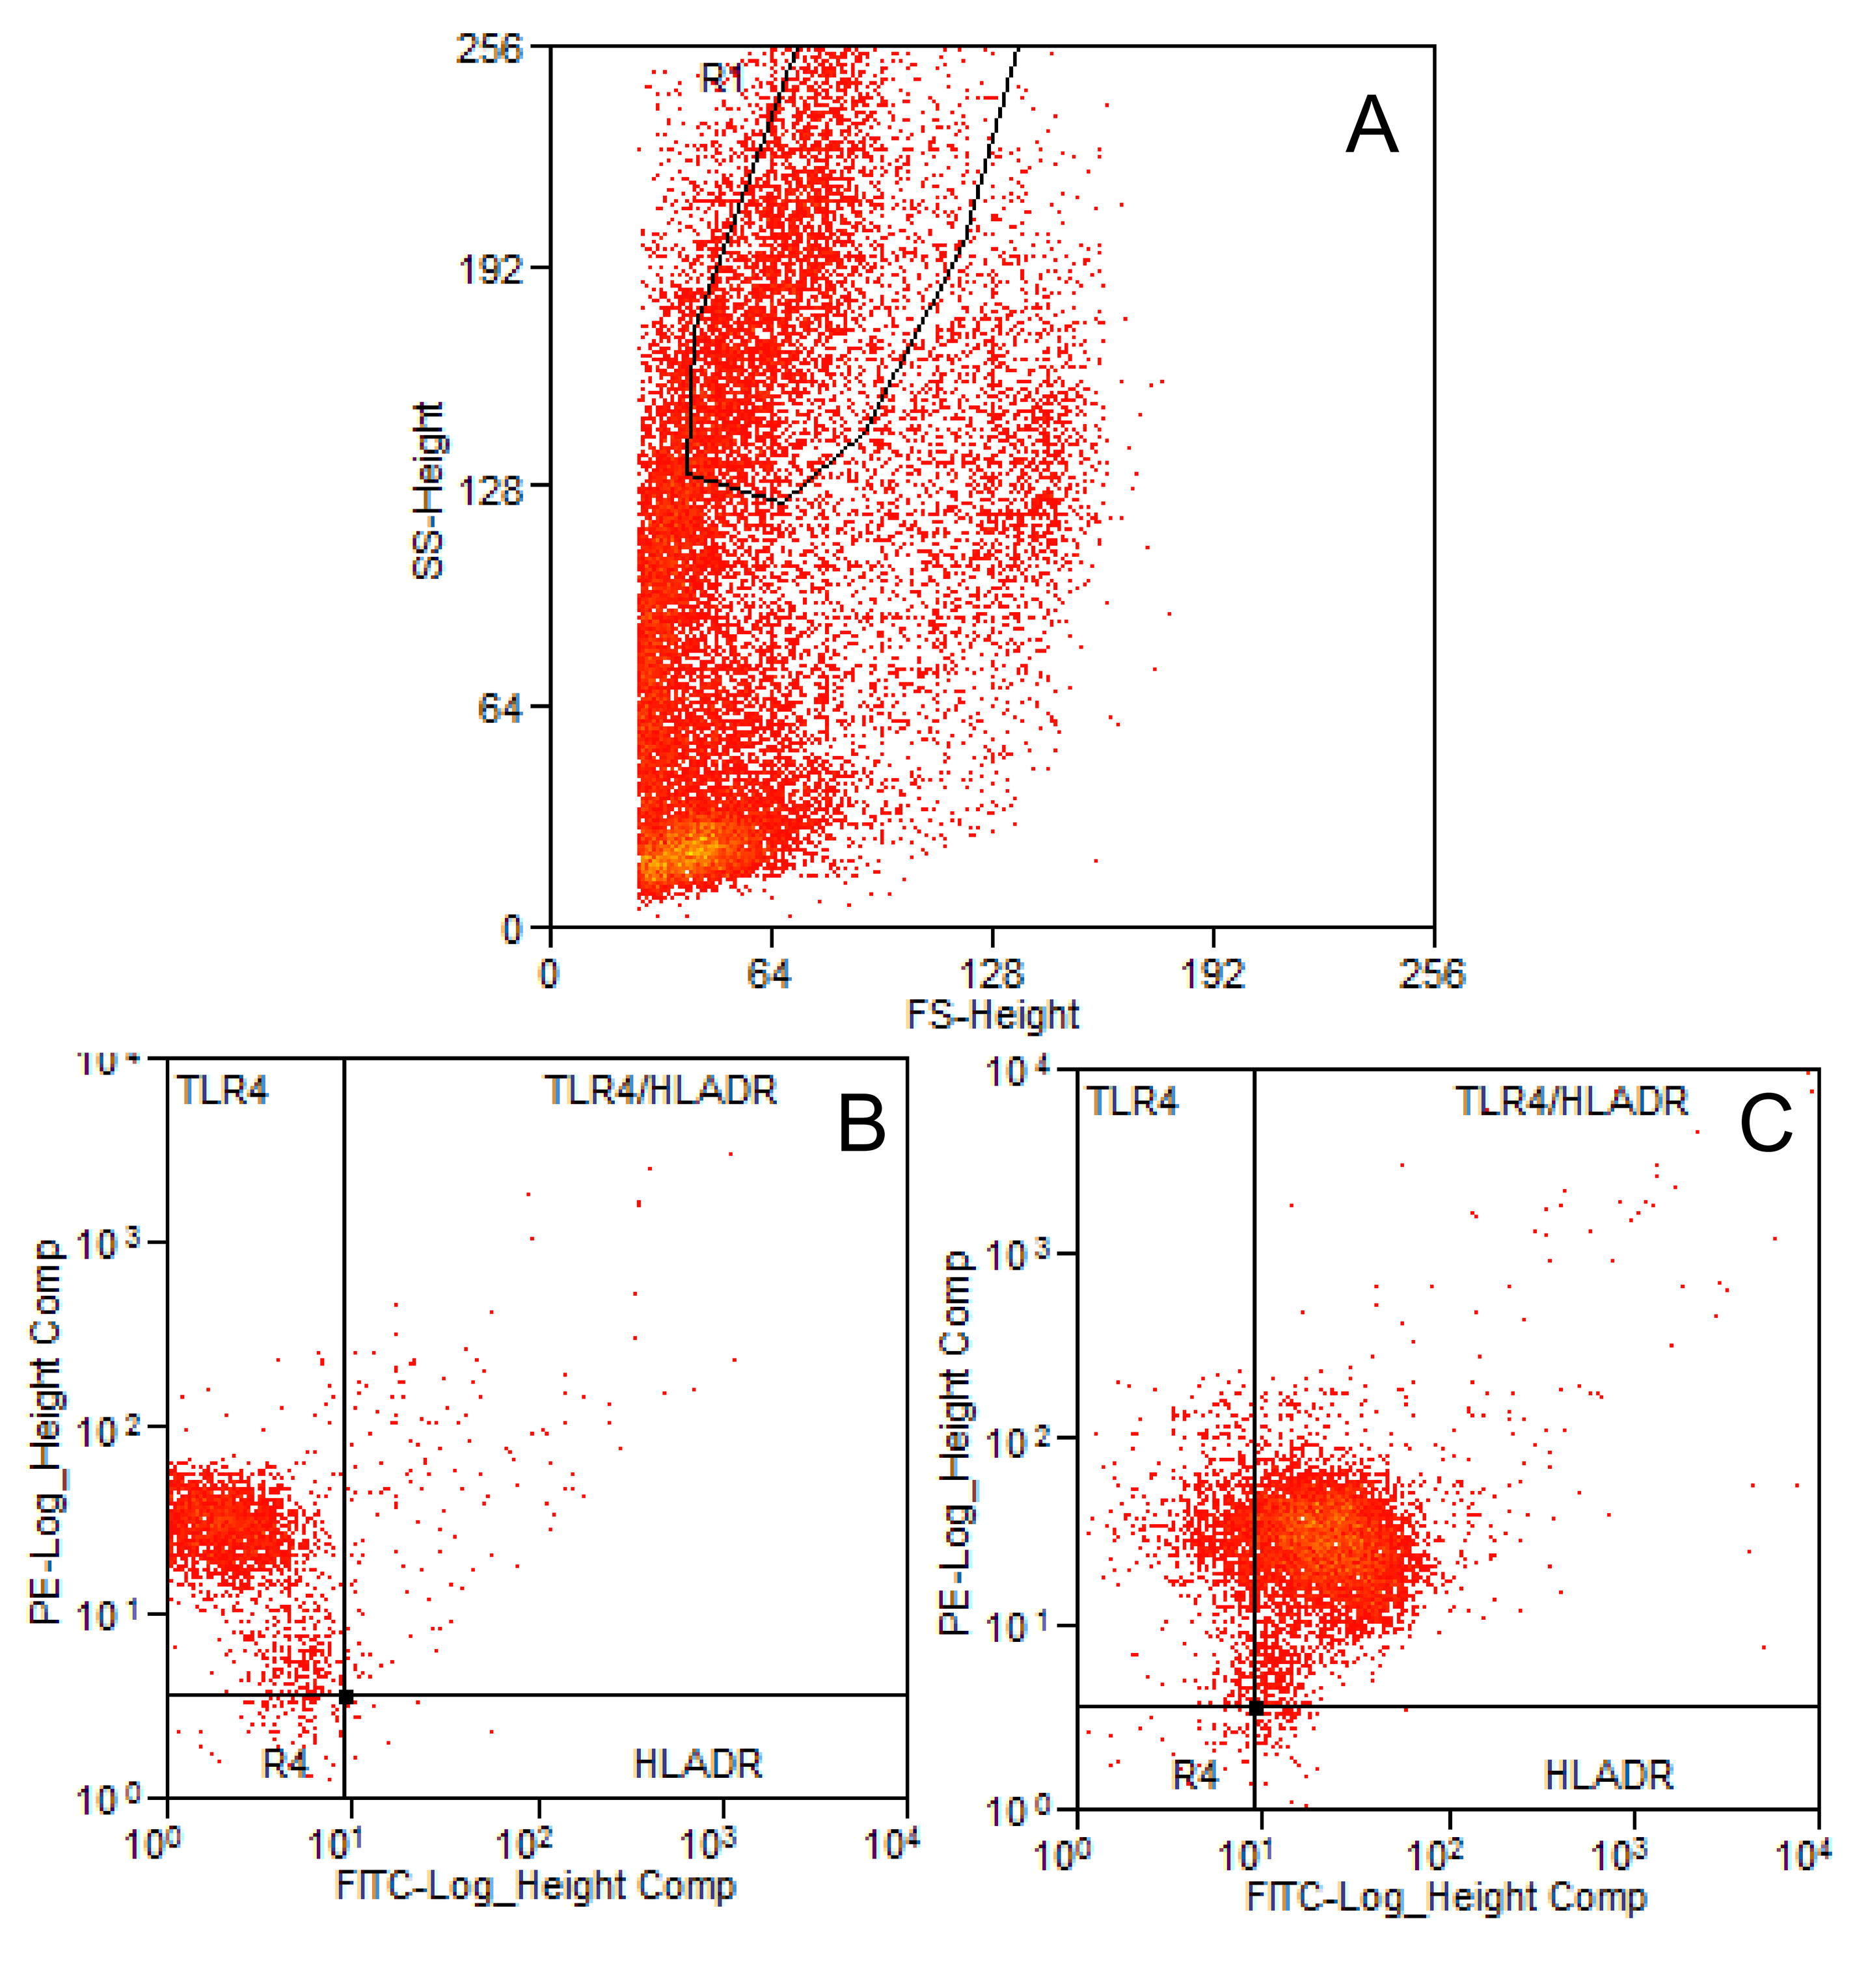
A
SS-Height
FS-Height
R1
B
C
TLR4
TLR4/HLADR
R4
HLADR
PE-Log_Height Comp
FITC-Log_Height Comp

Figure 1. Shows results from interpretation of flow cytometry. **A:** The PMNs were first selected based on logical gating on a forward scatter/side scatter plot. **B:** Cells challenged with LPS and not stimulated with GM-CSF. The population of cells shows TLR4 positive staining only. **C:** Cells challenged with LPS and stimulated with the low concentration of GM-CSF are shifted to the right, demonstrating an increase in HLADR expression.

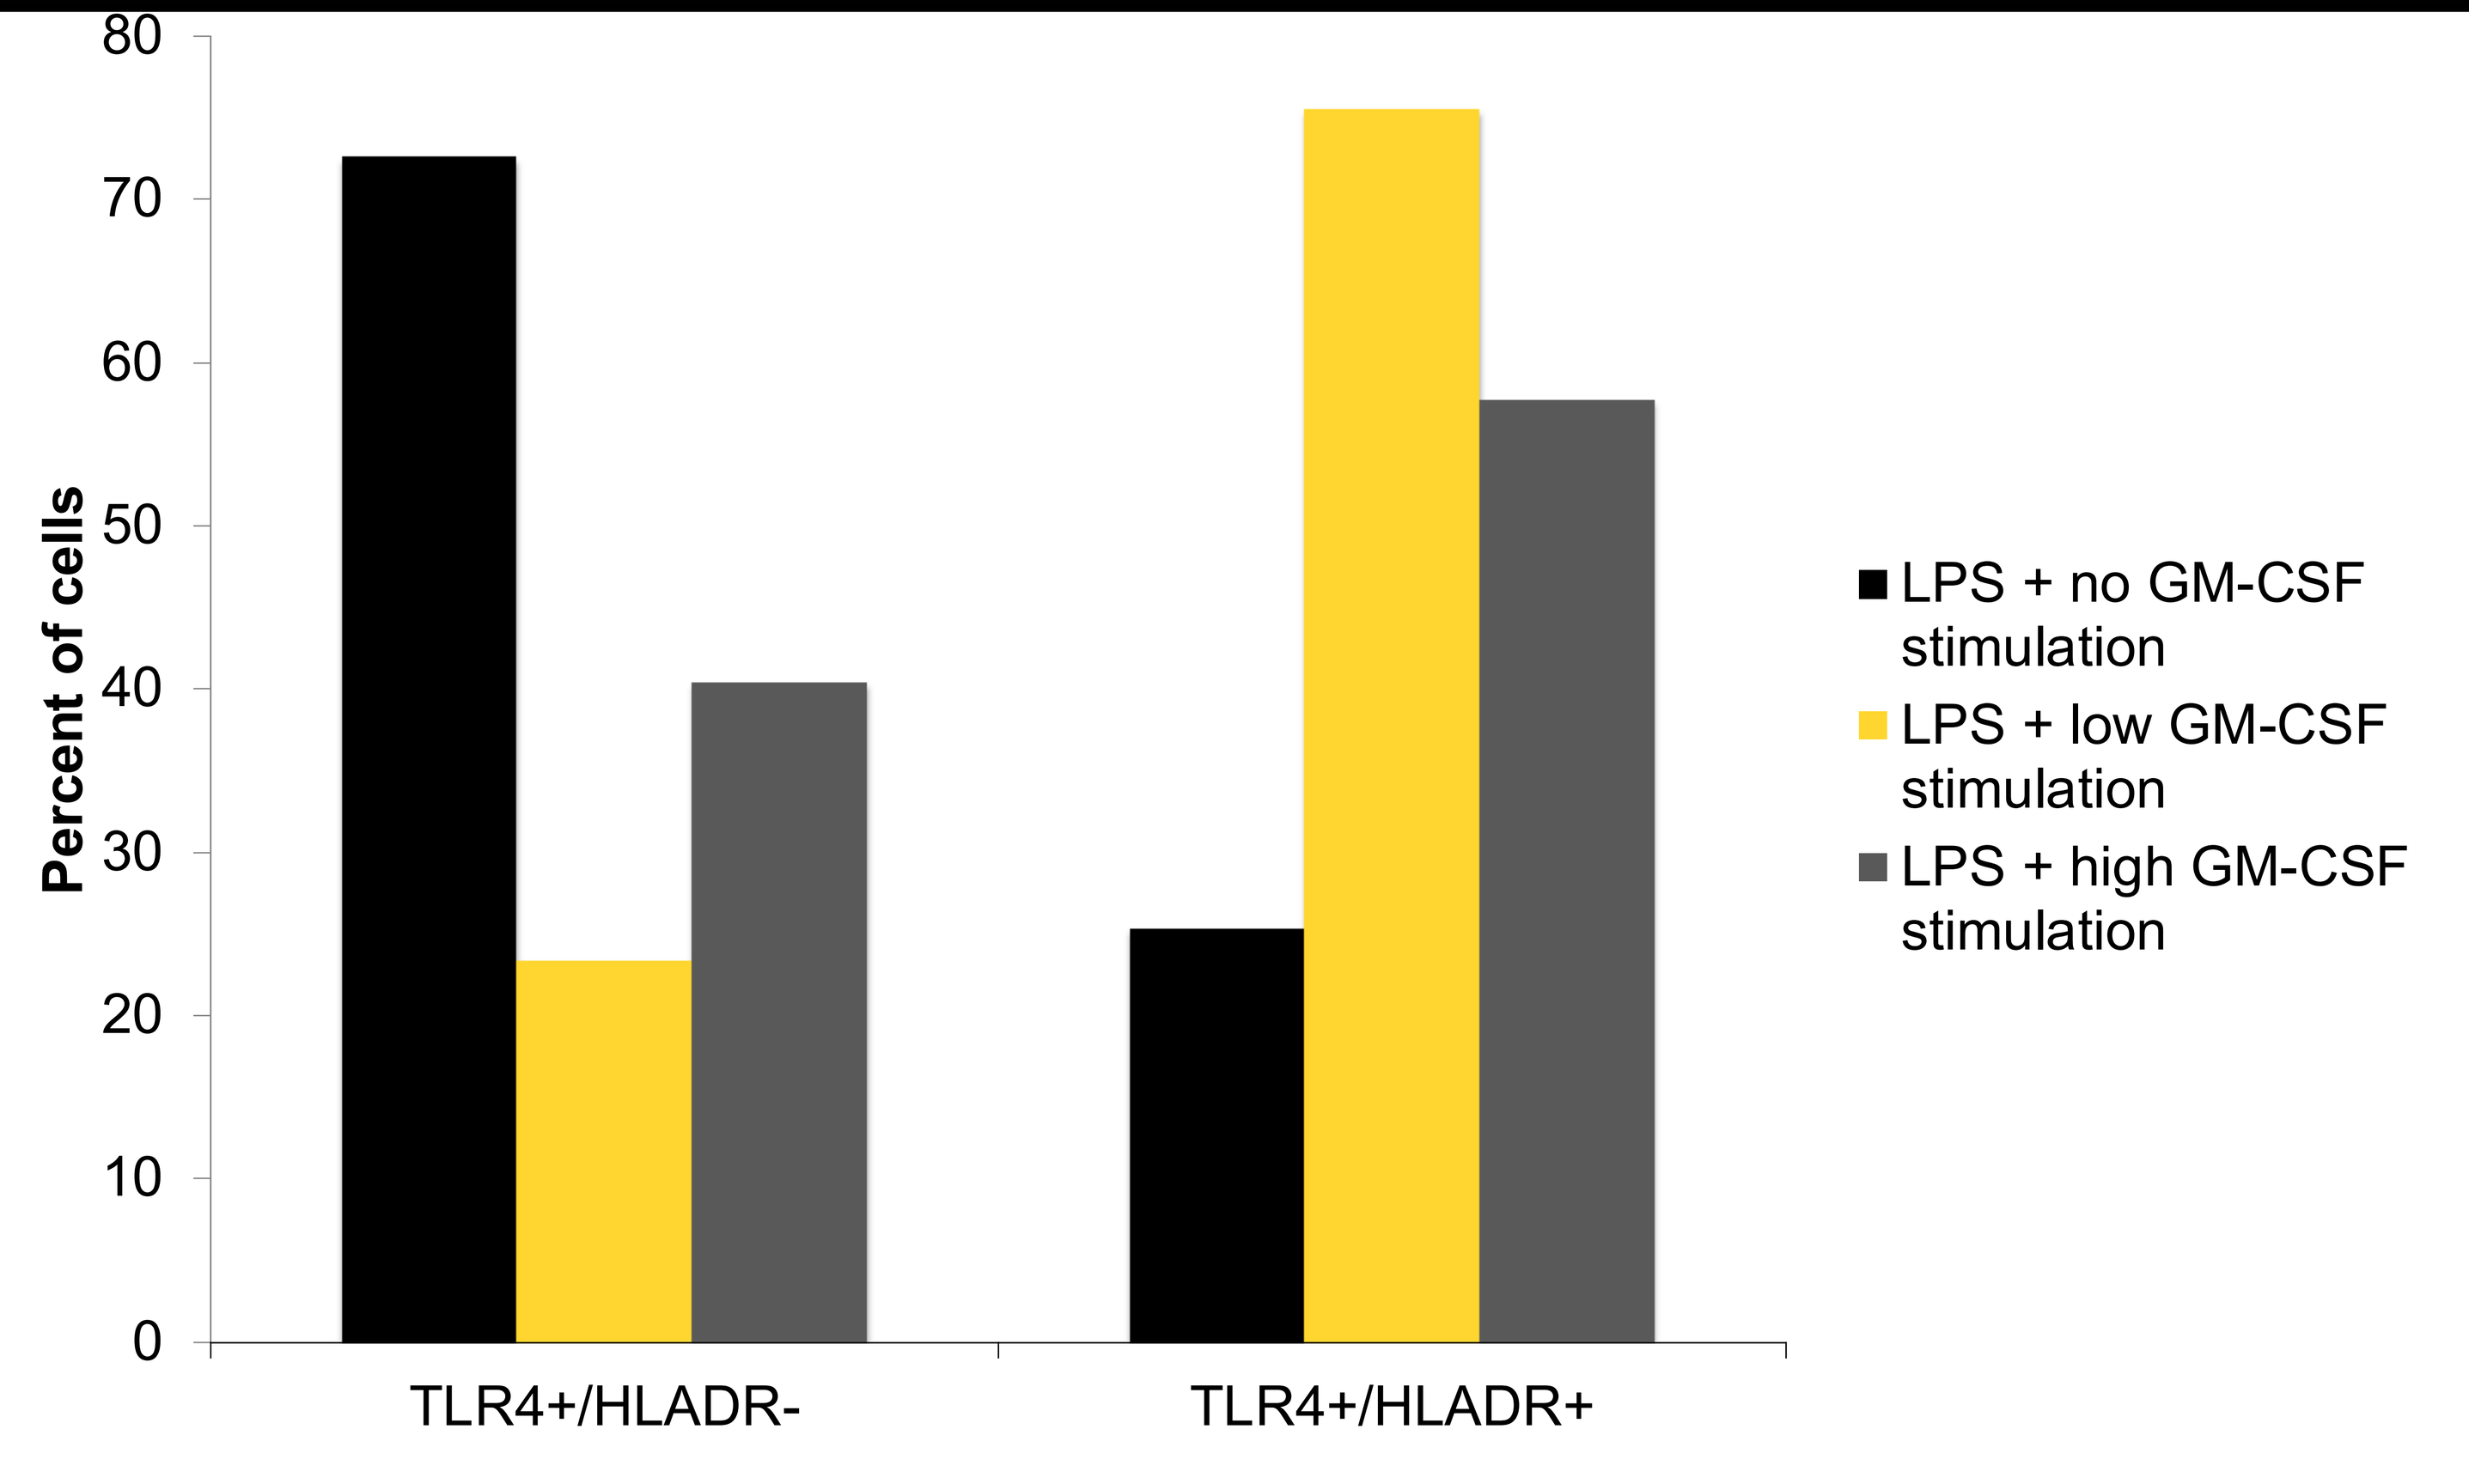
Percent of cells
TLR4+/HLADR-
TLR4+/HLADR+
LPS + no GM-CSF stimulation
LPS + low GM-CSF stimulation
LPS + high GM-CSF stimulation

Figure 2. Expression of TLR4 and HLADR on PMNs challenged with LPS and stimulated with no GM-CSF (control), a low concentration of GM-CSF, and a high concentration of GM-CSF. The stimulated cells, both with high and low concentration of GM-CSF, show increased expression of HLADR.

Results

Cell viability

The mean±SD viability was 95.6±1.2% indicating that GM-CSF did not alter cell viability compared to the control solution.

HLADR and TLR4 expression on PMNs

TLR4 and HLADR double-positive cell numbers increased in healthy dogs when treated with the low and high concentrations of GM-CSF (Figure 1 and 2).

Leukocyte cytokine production capacity

We have collected supernatant for cytokine production analysis and it is being stored at -80° C until analysis.

Evaluation of blood from dogs with cancer

Data collection is ongoing.

Conclusions

- GM-CSF did not cause cytotoxicity at a rate above 12%, which is acceptable viability for the study.

- Low and high concentrations of GM-CSF increases the number of TLR4/HLADR double positive PMNs in healthy dogs. While we do not yet have results, we hope to find that GM-CSF increases production of TNF-α.

- The next step is to perform these assays on blood from dogs with neoplasia and if the results are consistent, GM-CSF could be a useful treatment for dogs with neoplasia. Ultimately, the goal of the study is to apply our results in a clinical trial in tumor-bearing dogs with the hope of improving outcomes in dogs with neoplasia.

References

1. Vail DM, MacEwen EG. Spontaneously occurring tumors of companion animals as models for human cancer. Cancer Invest 2000;18:781-792.
2. Fleming JM, Creevy KE, Promislow DE. Mortality in north american dogs from 1984 to 2004: an investigation into age-, size-, and breed-related causes of death. J Vet Intern Med 2011;25:187-198.
3. Vaughan A, Johnson JL, Williams LE. Impact of chemotherapeutic dose intensity and hematologic toxicity on first remission duration in dogs with lymphoma treated with a chemoradiotherapy protocol. J Vet Intern Med 2007;21:1332-39.
4. Gosselin EJ, Wardwell K, Rigby WF, Guyre PM. Induction of MHC class II on human polymorphonuclear neutrophils by granulocyte/macrophage colony-stimulating factor, IFN-gamma, and IL-3. J immunol 1993;151(3):1482-90.

Funding and Acknowledgments

- A grant from Merial, a Sanofi Company
- University of Missouri, Comparative Internal Medicine Laboratory
- Thank you to Juliana and Dr. Zhang for your help in the lab and to Dr. DeClue for your support throughout this project.